Early Transendocardial Autologous Bone Marrow Injection Of Bone Marrow Derived Mononuclear Cells Following Ischemic Myocardial Events (the Alster - Helix Phase I Study)

Martin W Bergmann1, Christian Heeger2, Kai Jaquet2, Christoph Boosfeld3, Peter Altman3
1 Cardiologicum Hamburg, Hamburg, Germany, 2 Abt. Kardiologie, Ak StGeorg, Hamburg, Germany, 3 BioCardia, San Carlos, CA

Abstract

Background: There has been no clinical experience in an acute setting reported using a fluoroscopically guided system for intramyocardial delivery of cell therapy.
Objectives: To assess safety and efficacy of fluoroscopically guided intramyocardial transendocardial delivery of cell therapy less than 45 days after onset of acute myocardial infarction.
Methods: Patients with symptomatic heart failure following myocardial infarction (NSTEMI, STEMI) received transendocardial application of autologous bone marrow-derived mononuclear cells (BMC) 2-4 weeks after the acute event. A cardiac MRI was mandatory defining areas of less than 5mm wall thickness (no treatment) and of microvascular obstruction (treatment targets).
Results, Patients (n=9) with LV ejection fraction (EF) of 40.1± 8.0% and NYHA Class ≥II had autologous bone marrow cell preparation performed on site employing a closed-loop system. Cells delivered were 1.3± 1.3% CD34+, 1.68± 1.58% CD117+, 0.24± 0.16% CD133+, and 0.28± 0.40% CD90+. Each patient received Helix transendocardial injection of BMC into the infarction border zone 28±13 days following successful interventional revascularization. Delivery procedure took 32.1±11.7 minutes to perform 9± 2 deliveries of in total more than 1.5×10^8 cells/patient around the infarcted zone; There were no treatment emergent adverse events, no MACE, no pericardial effusions, and no arrhythmias in any procedure. Endpoints derived from comparisons of baseline vs. twelve month follow-up showed improvements of NYHA class (2.6±0.5 to 1.3 ± 0.5, p = 0.0002), BNP levels (362.1± 340.4 to 58.9 ± 45.9 ng/l, p = 0.036), and LV EF transthoracic echocardiography measurements (40.1±8.0 to 47.4 ± 9.0%, p = 0.02).
Conclusions: Results support that transendocardial intramyocardial injection of BMC using Helix can be used safely in patients with symptomatic heart failure following acute ischemic events.

Background

Heart failure is in need of new therapies aimed at preventing or reversing cardiac remodeling and at enhancing cardiovascular regeneration. This past decade has seen the emergence of protein, gene, and cell therapies to enhance cardiovascular regeneration.

The clinical safety and efficacy of intramyocardial delivery of autologous bone marrow-derived cells for the treatment of ischemic heart failure and chronic myocardial ischemia in twenty-three randomized controlled trials have been recently reviewed by the Cochrane Collaboration and the results are overall positive. Significant clinical benefit with a reduction in both mortality and rehospitalization due to heart failure were observed at long term follow-up in patients with chronic ischemic heart disease and heart failure. Significant improvements in heart failure symptoms measured by New York Heart Association Class (NYHA) and in functional capacity measured by left ventricular ejection fraction (LVEF), left ventricular end systolic volume (LVESV), at short-term and long-term follow-up were also observed.

No trials have been performed in an acute infarct setting using fluoroscopically guided transendocardial delivery due to hypothetical safety concerns that damage to friable myocardium resulting in perforation and tamponade could occur. However, it is well recognized that intramyocardial delivery can be 18 fold as efficient at delivering these cells to the myocardium as an intracoronary delivery route of administration (1).

Methods

Immediately prior to the catheterization procedure, autologous bone marrow was aspirated from the iliac crest, and concentrated as described previously (2). Cell samples were reserved pre and post concentration for measurement of surface markers using flow cytometry to delineate cellular composition with details on lymphocyte, granulocyte, monoctye, CD34+, CD117+, CD133+, and CD90+ progenitor stem cells.

Patients (n=9) with LV ejection fraction (EF) of 40.1± 8.0% and NYHA Class ≥II received Helix transendocardial injection of BMC into the infarction border zone 28±13 days following successful interventional revascularization. All patients received a cardiac MRI prior to intervention to determine wall thickness and baseline ejection fraction, which was required to be < 45%.

After the fluoroscopy was positioned at the appropriate magnification, only the C arm was rotated to maintain registration of the anatomy with the screen overlays. In addition to the endocardial border, areas of reduced or absent contraction were demarcated on the overlay and the target area for treatment. Infarct territories with LV wall thickness thinner than specified in the instructions for use for the Helix transendocardial delivery catheter were not be injected.

An 8 French Morph deflectable guide catheter (BioCardia, San Carlos, CA) was connected to the cardiac catheterization manifold with an in-line hemostatic adapter, flushed with heparinized saline solution (2000 units/liter), then inserted through the arterial sheath, and advanced over a 0.035” J-tipped angiographic guidewire to the ascending aorta. The guidewire was advanced to the left ventricle retrograde across the aortic valve; then, the deflectable guiding catheter is advanced over the wire retrograde across the aortic valve and into the left ventricular chamber, aspirated and flushed.

Methods cont’d

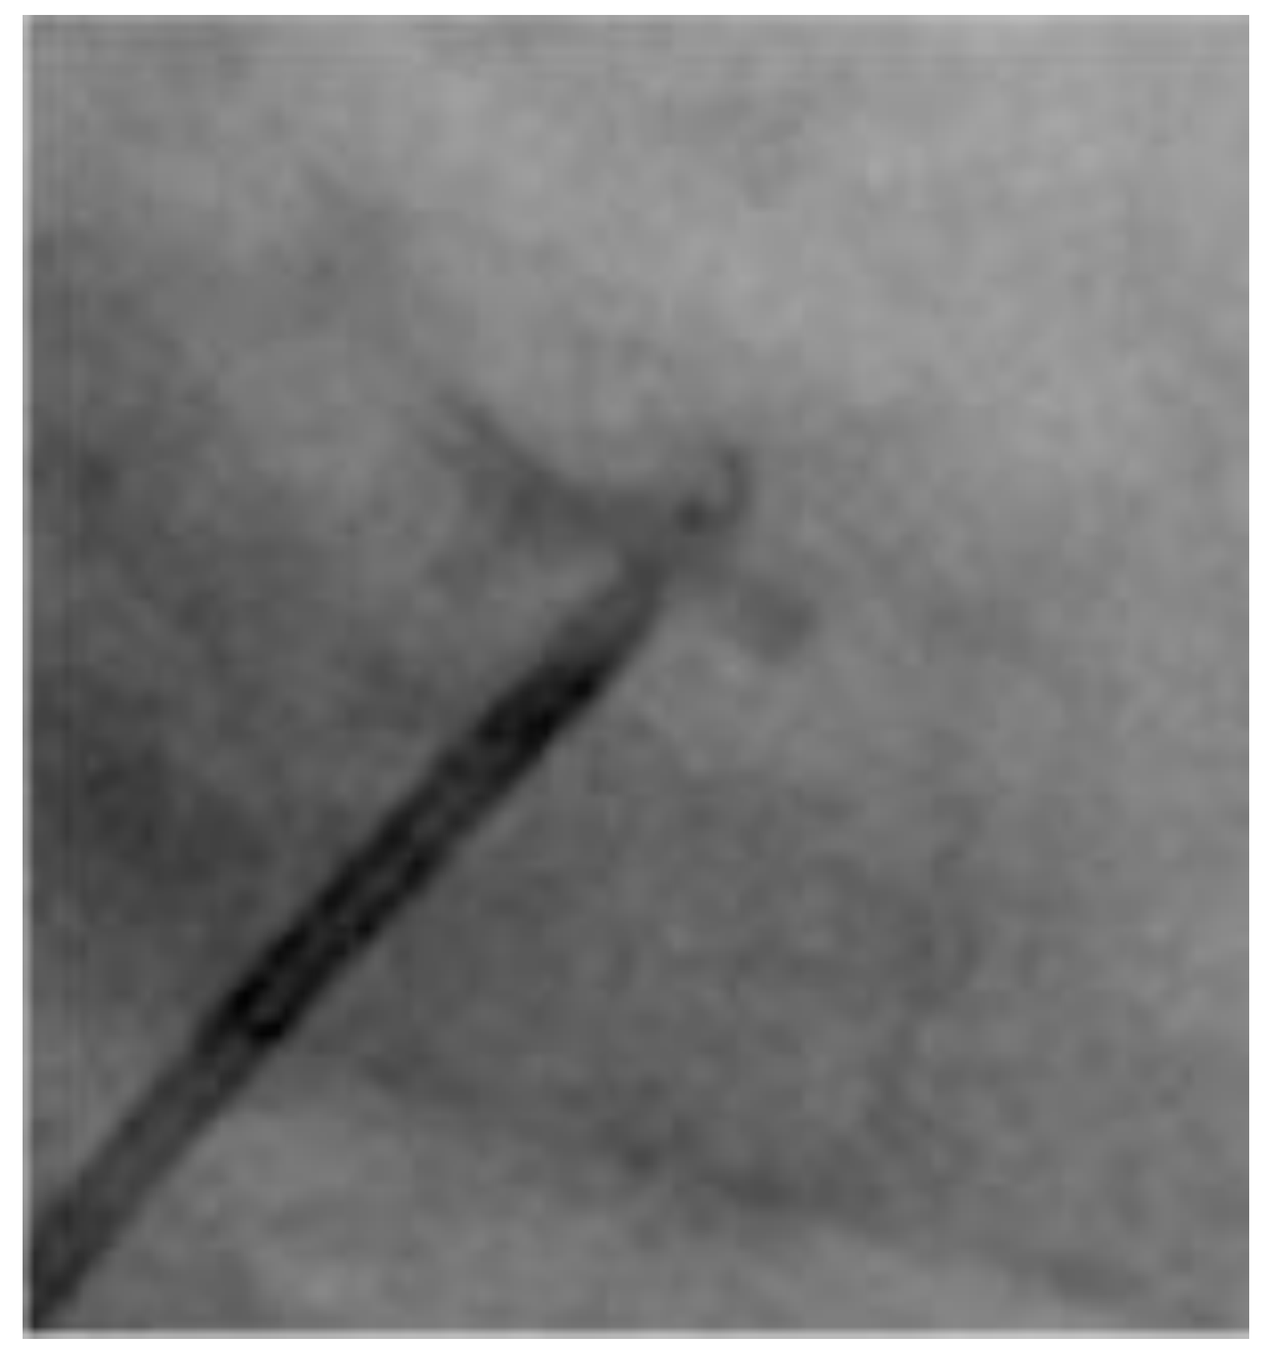

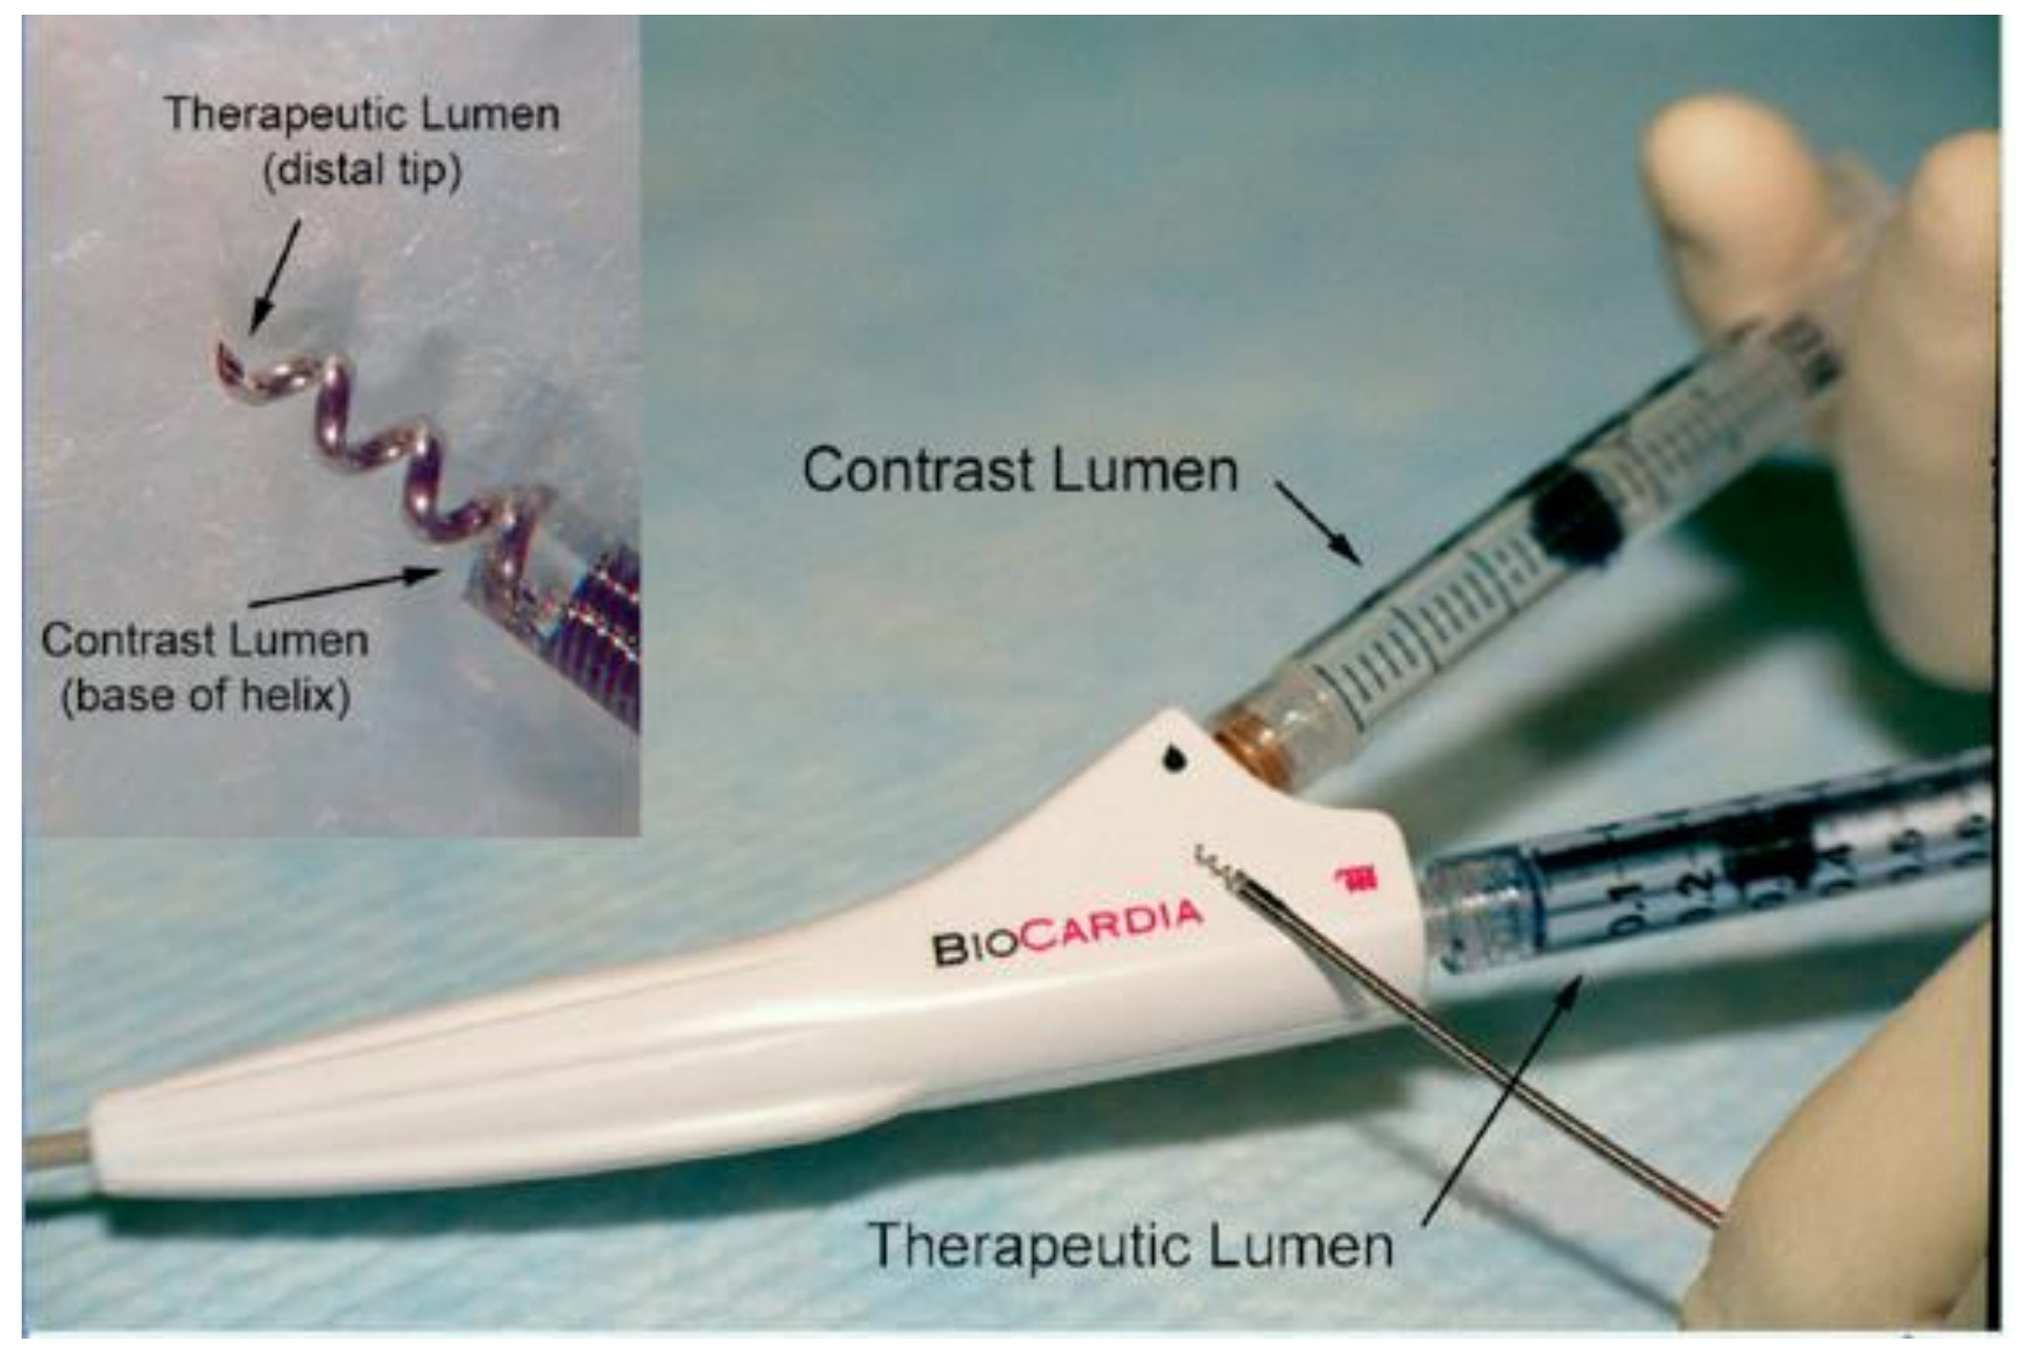
Therapeutic Lumen
(distal tip)
Contrast Lumen
(base of helix)
Contrast Lumen
BIOCARDIA
Therapeutic Lumen

Figure 1 (above left) shows intramyocardial engagement with contrast from base of Helix transendocardial biotherapeutic delivery system clearly marking the endocardial border. Helix handle and distal end shown in Figure 2 (above right).

Results

All delivery procedures were performed safely with no adverse events. There were no arrhythmias or pericardial effusions. Delivery procedure took 32.1±11.7 minutes to perform 9± 2 deliveries totaling 150×10^6 bone marrow mononuclear cells/patient around the infarcted zone. Primary and secondary endpoints were derived from comparisons of baseline vs. twelve month follow-up showing improvements of NYHA class (2.6±0.5 to 1.3 ± 0.5, p = 0.0002), BNP levels (362.1± 340.4 to 58.9 ± 45.9 ng/l, p = 0.036), and LV EF transthoracic echocardiography measurements (40.1±8.0 to 47.4 ± 9.0%, p = 0.086); calculated on an intra-individual basis EF improved by 9.3±2.5%, p=0.054.

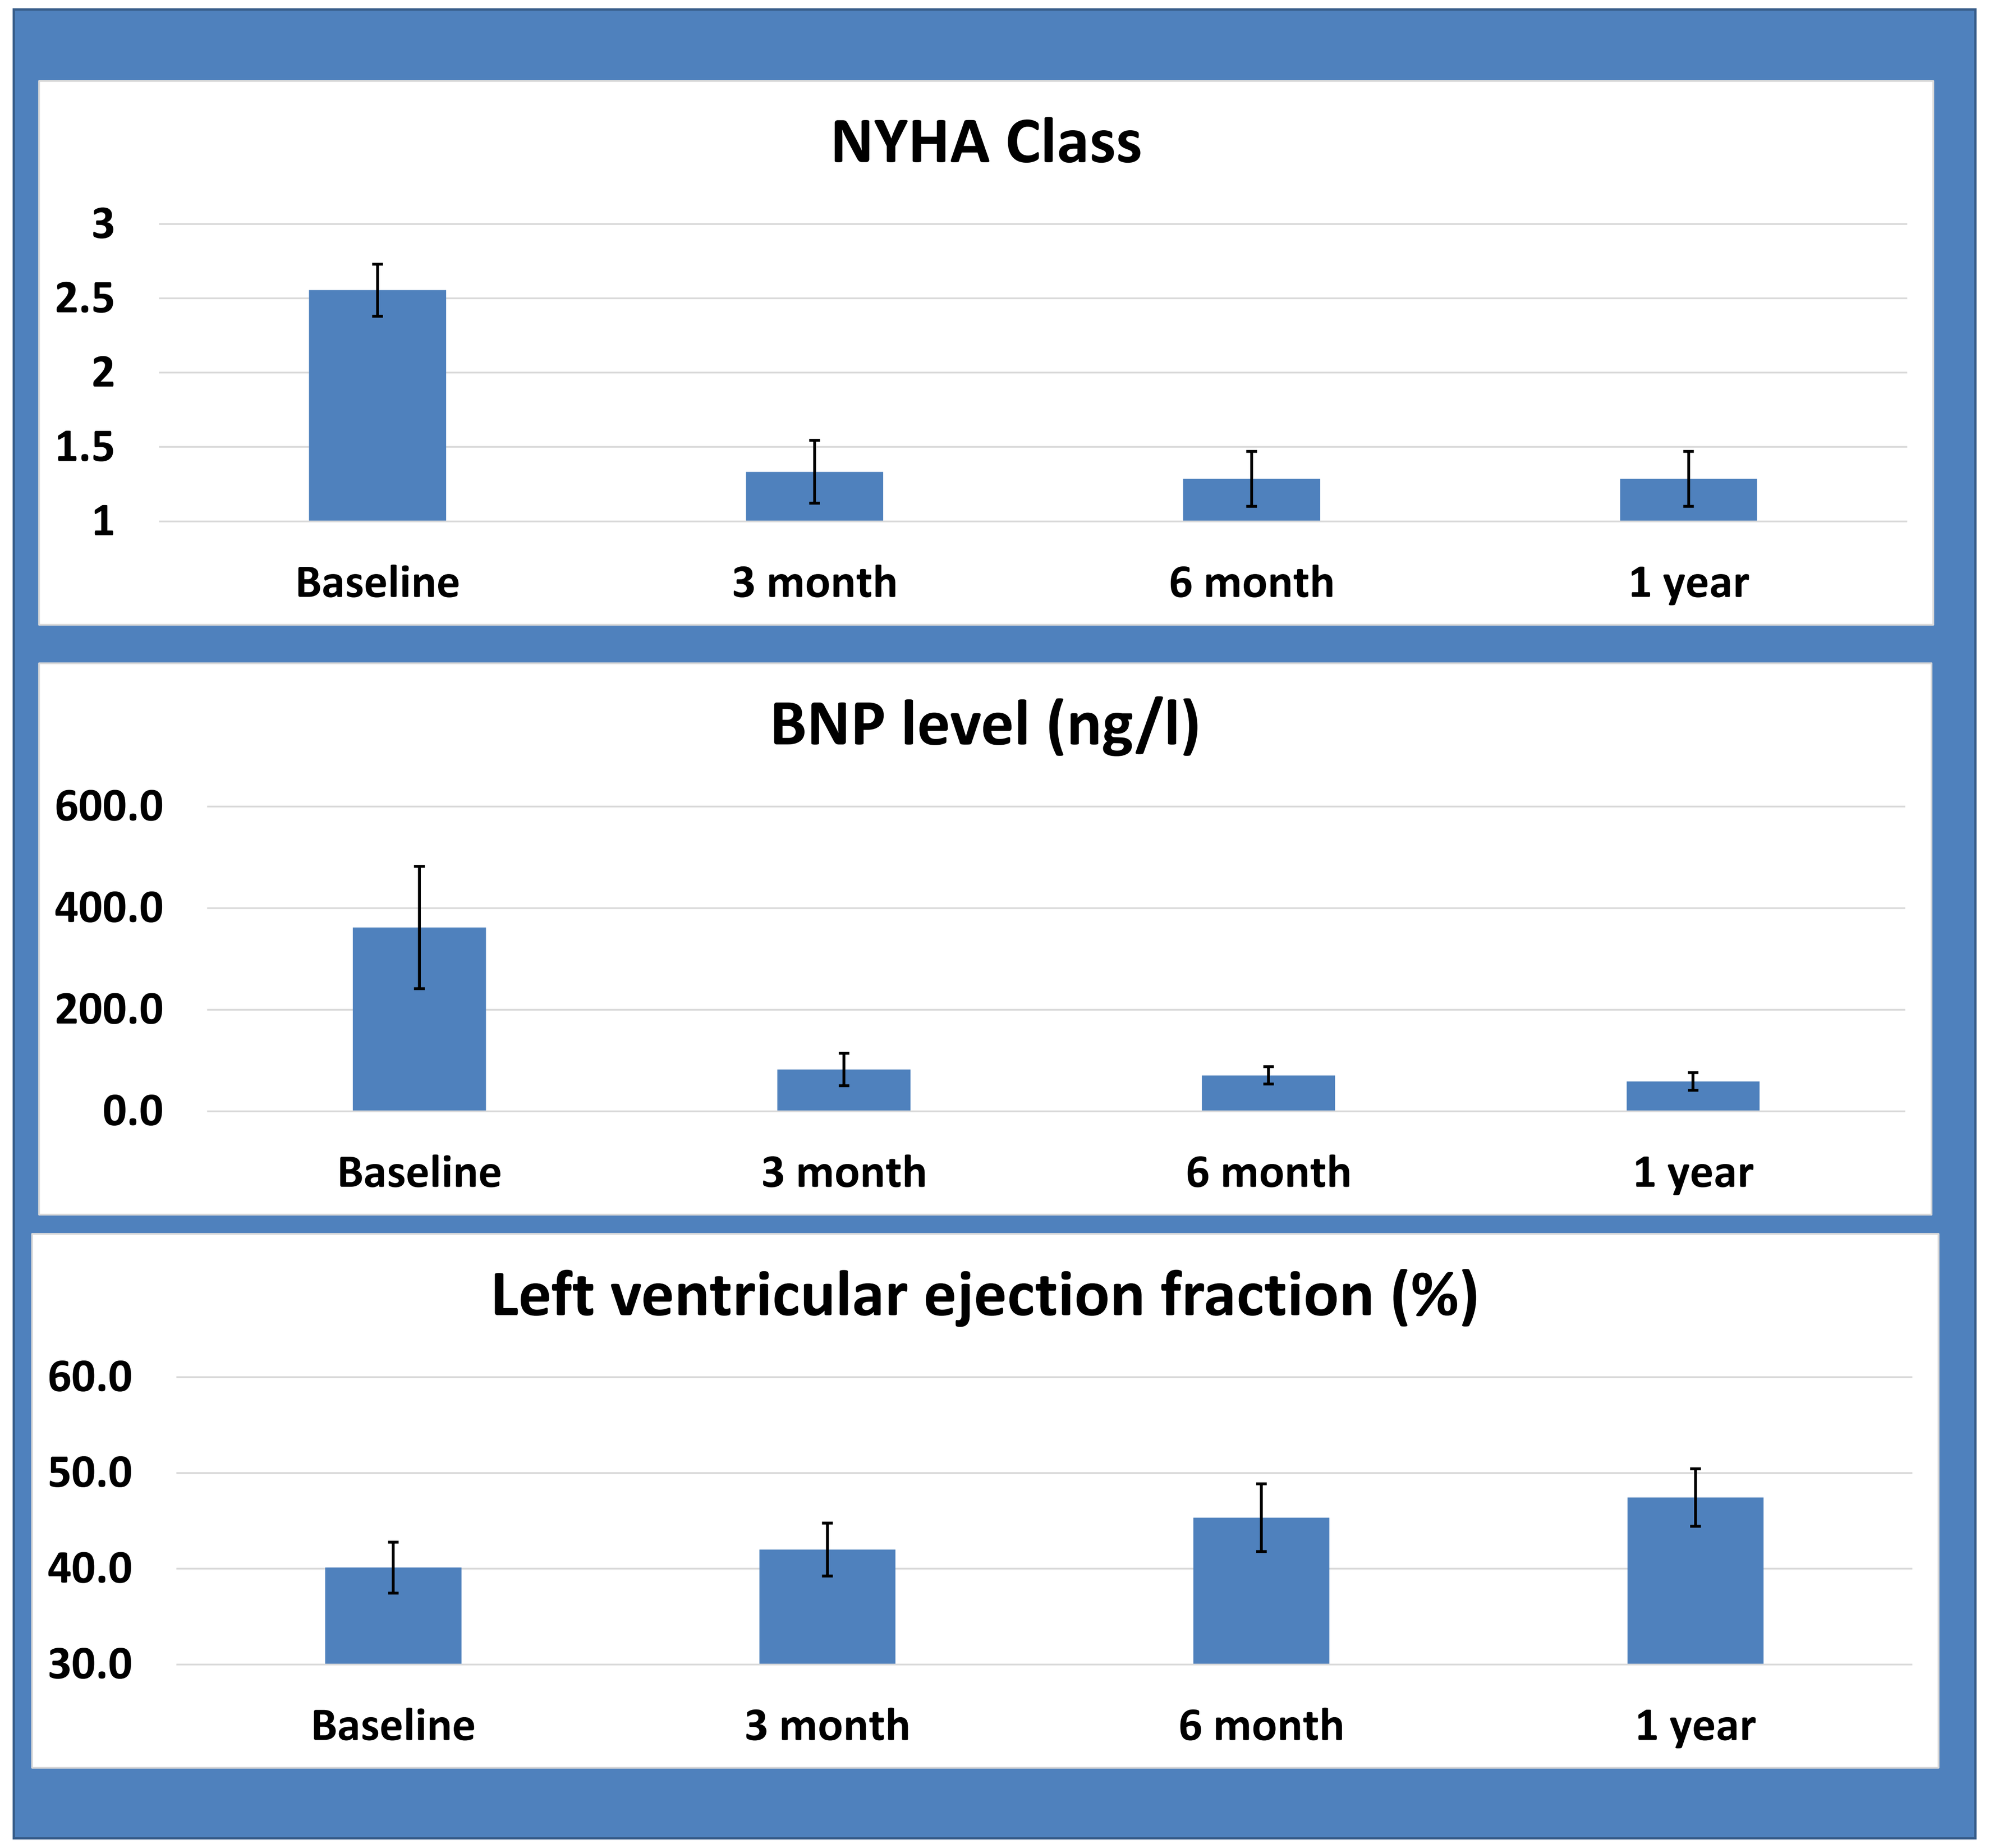
NYHA Class
3
2.5
2
1.5
1
Baseline
3 month
6 month
1 year
BNP level (ng/l)
600.0
400.0
200.0
0.0
Baseline
3 month
6 month
1 year
Left ventricular ejection fraction (%)
60.0
50.0
40.0
30.0
Baseline
3 month
6 month
1 year

Discussion

Here we show for the first time that fluoroscopically guided intramyocardial delivery can be performed safely in patients with symptomatic heart failure following acute ischemic events. The field has not explored this approach as there has been concern that intramyocardial delivery would be more dangerous in an acute setting due to the remodeling in the myocardium. Although a small series, the nine patients treated here were treated without incident and with compelling clinical results in this small safety cohort.

This work was initiated as recent literature shows us that different delivery approaches and systems can have drastically different efficiencies of delivery. Studies have shown that intramyocardial delivery with a Helix transendocardial delivery system, used in this series, delivers three fold more efficiently than straight needle intramyocardial delivery which in turn is six times more efficient than intracoronary artery delivery (1, 3).

This work supports that intramyocardial cell delivery approach used in small animal studies for this indication, can also be achieved in a minimally invasive fashion clinically. Such continuity of delivery methodology may enhance success of future development endeavors.

This work supports that advantages of targeted and highly efficient intramyocardial delivery that has been used to advance cell therapies in indications of chronic myocardial ischemia and heart failure may also have a role in a setting of acute infarction if the safety profile shown here can be extended to a larger population.

Coronary artery infusion in acute infarction historically has been shown to be safe, but does have complications. An extensive series performed with intracoronary delivery using the stop flow technique reported out on 775 patients treated showing a 1.7% treatment emergent MACE rate, excluding those associated with percutaneous coronary intervention, and a 0.5% mortality rate at 30 days (4).

The enhanced efficiency noted with intramyocardial delivery versus intra coronary artery infusion could be a key factor in enabling a therapy to be effective. Recent results in large trials in a similar clinical setting suggest that a more efficient intramyocardial delivery method could be important in a setting of acute infarction. The PreSERVE AMI trial which used intracoronary artery delivery of CD34+ cells had clearly reduced efficacy at lower doses (5) and might have benefited from an alternative delivery route. Other failed trials using intracoronary artery delivery might also have benefited from a clear assessment of efficiency of delivery with the selected route of administration.

Conclusion

The data suggest transendocardial injection of bone marrow derived progenitor and stem cells using Helix can be performed safely in patients with symptomatic heart failure following acute ischemic events. These data of a well-characterized, small cohort suggest efficiency compared to routine treatment. Further studies are required to confirm this work.

References

(1) Wong Po Foo C , Ikeno F, Altman PA, Rouy DB, Quantifying therapeutic cell retention in the heart to compare three routes of local delivery: Transendocardial Intramyocardial Injection, Transepicardial Intramyocardial Injection, and Intracoronary Artery Infusion, 8th International Conference on Cardiovascular Cell Therapy 2013.

(2) C. Heeger et al; EuroIntervention 2012 ALSTER stem cell trial

(3) Vrtovec B et al; Cell Comparison of Transendocardial and Intracoronary CD34+ Cell Transplantation in Patients With Nonischemic Dilated Cardiomyopathy

(4) Zeiher AM, Safety of the stop-flow technique for intracoronary cell administration: a single-center experience in 775 consecutive procedures, TCT, Miami Beach, October 2012.

(5) Quyyumi AA, PReSERVE AMI, Late Breaking Clinical Trials, American Heart Association 2014.

04453-A MKT